Healthcare Resource Utilization Trends for Patients with Inflammatory Bowel Disease and Iron Deficiency Anemia: A Real-World Claims Analysis

Cody Madsen, PharmD Candidate[1,2]; Alyssa Tutino, PharmD[3]; Christopher Diehl, PharmD, MBA, BCACP[3]; Eric Geisler[3]; Justin Bender, BS[3]

1. University of Utah College of Pharmacy, Salt Lake City, UT 2. Academy of Managed Care Pharmacy (AMCP) Foundation / Pfizer Managed Care Intern 3. Excellus BlueCross BlueShield, Rochester, NY

Background

- Inflammatory bowel disease (IBD) affects approximately 3 million adults in the United States and is associated with substantial healthcare resource utilization.[1-3]
- Iron deficiency anemia (IDA) is one of the most common extraintestinal complications of IBD and often persists in patients receiving advanced disease-modifying therapies.[4,5]
- American Gastroenterological Association (AGA) and European Crohn's and Colitis Organisation (ECCO) guidelines recommend intravenous (IV) iron for patients with moderate to severe IDA, for patients intolerant of oral iron, and for patients with active inflammation limiting absorption.[5,6]
- Published series report that anemia in IBD is frequently undertreated. Claims-based data describing IV iron use among IBD patients already receiving disease-modifying therapy remain limited, and this analysis was undertaken to help address that gap.[4-6]

Study Objectives

The primary objective of this study is to compare IBD-related healthcare resource utilization (HCRU) between patients with IBD and IDA who receive IV iron and those who do not.

The secondary objectives include:

- Comparison of adherence to IBD therapy between groups, measured as medication possession ratio (MPR)
- Comparison of corticosteroid utilization across the pharmacy and medical benefits

Methods

- A retrospective, observational cohort analysis was conducted using medical and pharmacy claims from a regional health plan in New York State.
- Members 4 months of age and older were included if they had: (1) a diagnosis of IBD, defined as Crohn's disease or ulcerative colitis; (2) a diagnosis of IDA; (3) evidence of disease-modifying therapy; and (4) continuous enrollment for 6 months before and after the index date.[11]
- Members were stratified by the presence of an IV iron claim under the medical benefit (IV iron utilizers vs IV iron non-utilizers), identified using Healthcare Common Procedure Coding System (HCPCS) codes.[7-10,12] The index date was the first IV iron claim with no prior IV iron claim in the preceding 6 months; index dates fell between January 2023 and December 2025.
- IBD-related utilization was defined as medical claims with a primary diagnosis of Crohn's disease (K50) or ulcerative colitis (K51), plus pharmacy claims for IBD therapies (aminosalicylates, anti-TNFs, interleukin inhibitors, anti-integrins, JAK inhibitors, and budesonide or rectal hydrocortisone). Claims coded primarily for IDA, including the IV iron infusions, fall outside this definition. Acute care is reported on an all-cause basis because few acute claims carried an IBD diagnosis.
- Observation windows differ by cohort. IV iron utilizers (n = 141) were observed for 13 months each: the 6 calendar months before the month of first infusion, the month of first infusion, and the 6 calendar months after. Claims were drawn from July 2022 through June 2026. Claims dated the same day as the index infusion were excluded from both periods.
- Members without an IV iron claim (n = 551) have no index event and were observed across the fixed 36-month calendar window from January 2023 through December 2025. The before and after comparison therefore applies to IV iron utilizers only.
- Healthcare professional (HCP) administered drugs were limited to agents with an approved indication for Crohn's disease or ulcerative colitis, together with IV iron.
- Members in the top and bottom 5% of total cost were removed from each cohort.
- Per member per month (PMPM) values use each cohort's own exposure.
- Members were classified as adherent if their MPR was 0.80 or greater over the measurement period.
- Corticosteroid and oral iron claims were counted across the full 2023 to 2025 window for both cohorts.
- Descriptive statistics were used for all outcomes.

Cohort selection

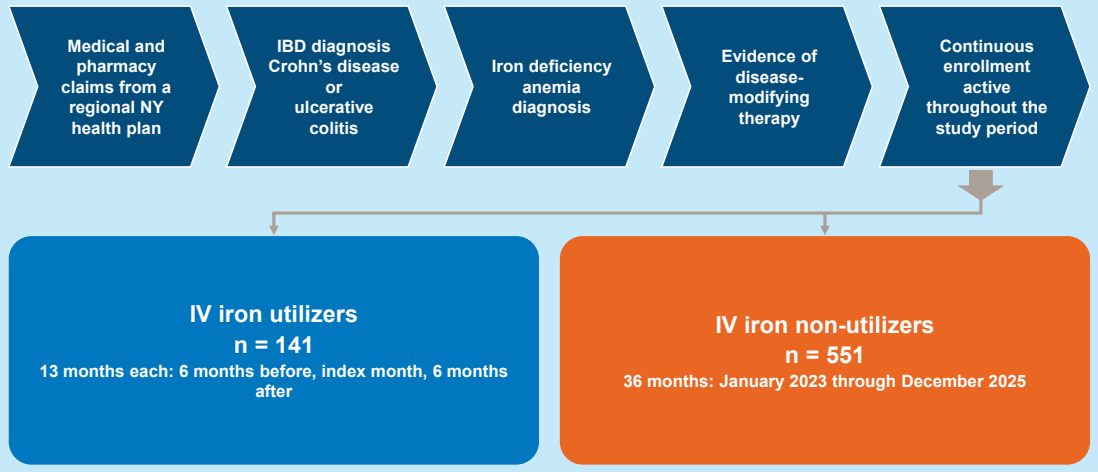
Medical and pharmacy claims from a regional NY health plan
IBD diagnosis Crohn's disease or ulcerative colitis
Iron deficiency anemia diagnosis
Evidence of disease-modifying therapy
Continuous enrollment active throughout the study period
IV iron utilizers
n = 141
13 months each: 6 months before, index month, 6 months after
IV iron non-utilizers
n = 551
36 months: January 2023 through December 2025

Charts and Tables

Table 1. Baseline Characteristics of the Study Population

Characteristic	IV iron n = 141	No IV iron n = 551
Demographics		
Mean age, years	47.7	54.3
Female, n (%)*	86 (61.0%)	346 (62.8%)
Male, n (%)*	55 (39.0%)	203 (36.8%)
Age group, n (%)*		
0-19	18 (12.8%)	22 (4.2%)
20-39	32 (22.7%)	105 (20.2%)
40-59	46 (32.6%)	162 (31.1%)
60-79	39 (27.7%)	189 (36.3%)
80+	6 (4.3%)	43 (8.3%)
Diagnosis, n†		
Crohn's disease	62	302
Ulcerative colitis	70	287

Table 2. IBD-Related Claims Utilization Before and After the Index Date, IV Iron Utilizers§

Category	6 months before	6 months after	Change
IBD-related allowed amount PMPM			
Inpatient	$349	$543	+55.6%
Outpatient	$921	$1,018	+10.5%
Professional	$347	$515	+48.4%
Pharmacy (IBD therapies)	$3,428	$3,986	+16.3%
HCP administered drugs PMPM			
IBD therapies and IV iron	$249	$488	+95.8%
IV iron products	*$2*	*$47*	–
IBD biologics	*$247*	*$441*	–

Figure 1. IBD-Related Claims Utilization per Member per Month by Setting and Cohort‡

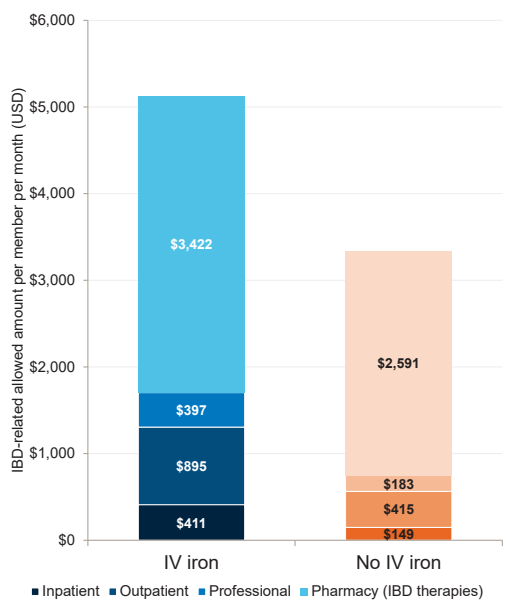
IBD-related allowed amount per member per month (USD)
$6,000
$5,000
$4,000
$3,000
$2,000
$1,000
$0
$3,422
$397
$895
$411
$2,591
$183
$415
$149
IV iron
No IV iron
Inpatient Outpatient Professional Pharmacy (IBD therapies)

Figure 2. Corticosteroid Claim Utilization by Benefit and Cohort

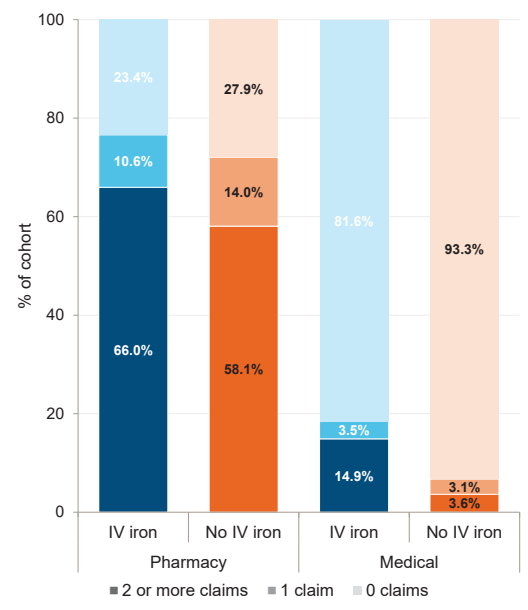
% of cohort
23.4%
10.6%
66.0%
27.9%
14.0%
58.1%
81.6%
3.5%
14.9%
93.3%
3.1%
3.6%
IV iron
No IV iron
IV iron
No IV iron
Pharmacy
Medical
2 or more claims 1 claim 0 claims

Figure 3. All-Cause Acute Care Claim Utilization by Cohort||

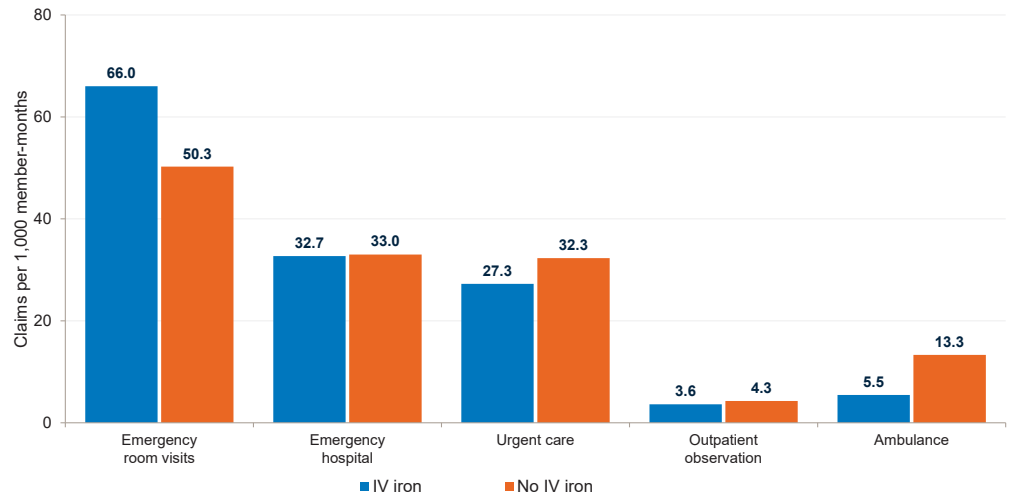
Claims per 1,000 member-months
66.0
50.3
32.7
33.0
27.3
32.3
3.6
4.3
5.5
13.3
Emergency room visits
Emergency hospital
Urgent care
Outpatient observation
Ambulance
IV iron
No IV iron

Figure 4. IBD Therapy Adherence by Drug Class and Cohort

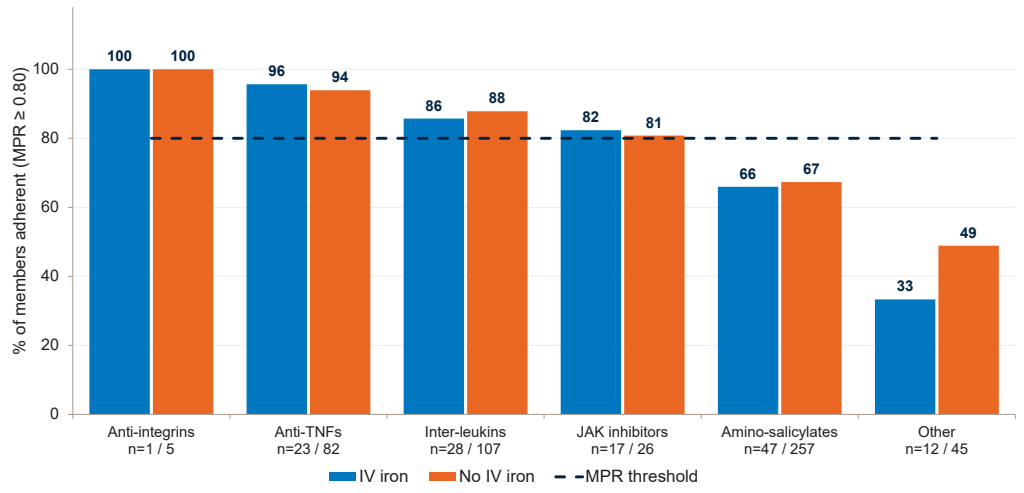
% of members adherent (MPR ≥ 0.80)
100
100
96
94
86
88
82
81
66
67
33
49
Anti-integrins n=1 / 5
Anti-TNFs n=23 / 82
Inter-leukins n=28 / 107
JAK inhibitors n=17 / 26
Amino-salicylates n=47 / 257
Other n=12 / 45
IV iron
No IV iron
MPR threshold

*Table 1: Sex was unknown for 2 IV iron non-utilizers, and age was available for 521 of 551 IV iron non-utilizers.
†Table 1: Diagnoses use the primary diagnosis on medical claims in each member's window, so a member may have both diagnoses
§Table 2: HCP administered drugs include IV iron and IBD-indicated agents regardless of claim diagnosis; the IV iron products and IBD biologics rows show each group's share of the increase.
‡Figure 1: Denominators are 1,651 member-months for IV iron utilizers and 16,812 (medical) and 17,892 (pharmacy) for IV iron non-utilizers. The Other setting (under $20 PMPM) is not shown.
||Figure 3: All-cause claims. Emergency room and emergency hospital claims are counted separately and are not added together.

Results

- **Study population:** A total of **141 IV iron utilizers** and **551 IV iron non-utilizers** met inclusion criteria. **IV iron utilizers were younger** (mean 47.7 vs 54.3 years), with a greater share under age 20 (12.8% vs 4.2%); sex distribution was similar (61.0% vs 62.8% female) (**Table 1**).
- **Primary objective, IBD-related HCRU:** Characterized using claims-based data, IBD-related utilization was higher among IV iron utilizers in every setting. Inpatient (**$411 vs $149**), outpatient (**$895 vs $415**) and professional (**$397 vs $183**) utilization per member per month were each **more than twice as high**, while IBD therapy pharmacy utilization was highest and remained closer ($3,422 vs $2,591) (**Figure 1**).
- All-cause acute care was mixed: IV iron utilizers had more emergency room visits (66.0 vs 50.3 per 1,000 member-months), similar emergency hospital claims (32.7 vs 33.0), and fewer urgent care and ambulance claims (**Figure 3**).
- Within IV iron utilizers, IBD-related utilization in the 6 months after the index date rose for inpatient (**+55.6%**), professional (+48.4%), pharmacy (+16.3%) and outpatient (+10.5%) care. Healthcare professional administered drugs rose **95.8%**; IBD biologics accounted for 81.3% of that increase and IV iron products for 18.7% (**Table 2**).
- **Secondary objective, adherence:** IBD therapy **adherence was equivalent between cohorts** and varied by drug class rather than by cohort, from 100% for anti-integrins (with 1 IV iron utilizer), 96% for anti-TNFs, 66% for aminosalicylates to 33% for other agents (**Figure 4**).
- **Secondary objective, corticosteroid utilization:** Use under the medical benefit was more frequent among IV iron utilizers: 14.9% vs 3.6% had 2 or more claims, 11.3 percentage points higher (**about 4 times as often**). Pharmacy benefit use was closer: 66.0% vs 58.1%, 7.9 percentage points higher (about 1.1 times as often) (**Figure 2**).

Discussion and Limitations

- **Primary objective:** IBD-related healthcare resource utilization was higher among IV iron utilizers, driven by medical care, which was **more than twice as high** across inpatient, outpatient and professional settings. IBD therapy spending differed less. Because acute care was mixed and rarely IBD-coded, the difference reflects ongoing IBD management rather than emergency use.
- **What drives the increase:** Within IV iron utilizers, most of the rise in professional administered drugs after the index date comes from IBD biologics rather than IV iron (**81.3% of the increase**), which suggests IBD therapy is started or intensified around the same time that iron-deficiency anemia is treated.
- **Unmeasured disease severity:** IV iron use may point toward **more severe IBD than claims capture**, because ICD-10 codes for Crohn's disease and ulcerative colitis do not grade severity. IV iron utilizers may have more active disease without a diagnosis that reflects it.
- **Age differences:** IV iron utilizers were younger than non-utilizers (mean age 47.7 vs 54.3 years), with the most notable being a greater proportion under 20 years of age (**12.8% vs 4.2%**) (Table 1). This may reflect differences in disease severity or treatment patterns by age. As the cohorts were not age matched, age-related differences may have contributed to the observed variation in IBD-related HCRU.
- **Adherence:** Greater adherence was observed for advanced therapies than for conventional treatments such as aminosalicylates, and this pattern was present in both cohorts, so **IV iron use is not a marker of better or worse adherence**.
- **Corticosteroid utilization:** Greater medical benefit use among IV iron utilizers **may reflect more active/severe disease**.
- **Oral iron use, an additional measure:** Pharmacy claims were reviewed to assess whether members received a trial of oral iron before IV iron. Most did not: 74.5% of IV iron utilizers and 81.9% of IV iron non-utilizers had no oral iron claim. This should be interpreted with caution, because oral iron is widely available over the counter and often purchased without a prescription. Claims data therefore likely underestimate true oral iron use, and prior oral iron trials may not be fully captured.
- **Outlier handling:** Members with extreme total costs were excluded from each cohort to avoid skewed cost estimates driven by atypical spending patterns.
- **Limitations:** IBD-related utilization relies on the primary diagnosis, so IBD care coded under a secondary diagnosis is missed, and IDA-coded claims, including the infusions, are excluded by design. As mentioned, oral iron is largely available over the counter, so use outside the pharmacy benefit cannot be observed and true oral iron use is understated. Differences in observation windows between cohorts may affect direct comparisons, despite normalization of outcomes per member per month or per 1,000 member-months. Findings are descriptive as **no inferential statistics were performed**. Findings reflect a single regional health plan and may not be generalizable.

Conclusions

- **Primary objective:** IBD-related healthcare resource utilization was higher among IV iron utilizers in every setting, with inpatient, outpatient and professional utilization each more than twice as high. Within IV iron utilizers, most of the increase in professional administered drugs after the first infusion reflects IBD biologics rather than the iron itself.
- **Secondary objectives:** Adherence to IBD therapy was equivalent between cohorts; the variation that did appear was between advanced and conventional therapy classes and was present in both groups. Corticosteroid utilization under the medical benefit was more frequent among IV iron utilizers.
- **Managed care implications:** These findings support monitoring IBD-related medical and professional administered drug spend in this population, reviewing site of care for infusion administration, and directing adherence support toward conventional oral maintenance therapy. IV iron use may also flag members with more severe IBD for care management outreach.
- **Next steps:** Further investigation is warranted to determine whether IV iron therapy may serve as a marker of IBD severity, given inconsistencies in ICD-10 coding practices. Electronic health record (EHR) data would be necessary to further evaluate this potential association.

References

1. Centers for Disease Control and Prevention. Inflammatory Bowel Disease (IBD). Atlanta, GA: US Department of Health and Human Services; 2024.
2. National Institute of Diabetes and Digestive and Kidney Diseases. Inflammatory Bowel Disease. Bethesda, MD: National Institutes of Health; 2024.
3. Crohn's & Colitis Foundation. The Facts About IBD. New York, NY: Crohn's & Colitis Foundation; 2024.
4. World Health Organization. Anaemia. Geneva, Switzerland: WHO; 2024.
5. Ko CW, Siddique SM, Patel A, et al. AGA Clinical Practice Guidelines on the Management of Iron Deficiency Anemia. Gastroenterology. 2020;159(3):1141-1151.
6. Dignass AU, Gasche C, Bettenworth D, et al. European consensus on the diagnosis and management of iron deficiency and anaemia in inflammatory bowel diseases. J Crohns Colitis. 2015;9(3):211-222.
7. American Regent, Inc. Venofer (iron sucrose) Injection Prescribing Information. Shirley, NY; 2024.
8. American Regent, Inc. Injectafer (ferric carboxymaltose) Injection Prescribing Information. Shirley, NY; 2024.
9. AMAG Pharmaceuticals. Feraheme (ferumoxytol) Injection Prescribing Information. Waltham, MA; 2024.
10. Pharmacosmos Therapeutics Inc. Monoferric (ferric derisomaltose) Injection Prescribing Information. Parsippany, NJ; 2024.
11. Centers for Disease Control and Prevention; Centers for Medicare & Medicaid Services. ICD-10-CM Official Guidelines for Coding and Reporting. Baltimore, MD: CMS; 2024.
12. Centers for Medicare & Medicaid Services. HCPCS Level II Code Set. Baltimore, MD: CMS; 2024.

Contact Info: cody.madsen@comcast.net | Poster presented at AMCP Nexus 2026, October 26-29, San Diego, CA | This research was completed as part of the 2026 AMCP Foundation Managed Care Internship, sponsored by Pfizer, Inc. | Special thanks to Tyson Thompson, PharmD, MBA, and Piao Ching, PharmD, of Pfizer, Inc., for their collaboration on this poster.